In Vivo Pooled Screening Platform for the Discovery of Optimized Chimeric Antigen Receptor (CAR) Design in T cells

Joshua Mace[1], Matthew Forsberg[2], Nina La Vonne Denne[3,4], Siqi Zhao[1], Khloe Gordon Wei[1], Jai Raman[1], Narendra Maheshri[1], Christian Capitini[2], Krishanu Saha[3,4], Shawdee Eshghi[1], Taeyoon Kyung[1]

[1] Ginkgo Bioworks, Boston, MA, [2] University of Wisconsin School of Medicine and Public Health, Madison, WI, [3] Department of Biomedical Engineering, University of Wisconsin-Madison, Madison, WI, [4] Wisconsin Institute for Discovery, University of Wisconsin-Madison, Madison, WI

Background

While Chimeric Antigen Receptor expressing T cells (CAR-T cells) have shown high response rates against hematological cancers in the clinic, they have shown limited efficacy against solid tumors due to lack of persistence and immunosuppression in the tumor microenvironment (TME). To address the TME, recent efforts have focused on discovering novel intracellular signaling domains (ICDs) with enhanced anti-tumor efficacy at scale. However, in vitro screens are inherently limited in mimicking the TME and other physiologically relevant aspects of adoptive cell therapy (ACT).

Here, we developed an in vivo pooled library screen approach alongside our existing in vitro platform to identify potent CAR designs in a CHLA-20 neuroblastoma xenograft model. We cloned a DNA-barcoded 10,000-member CAR ICD library into an anti-GD2 CAR backbone. GD2 CAR library-expressing primary T cells were injected into tumor-bearing mice at >100x library coverage, and spleens and tumors were collected 2 weeks post-ACT. Sequencing genomic DNA quantified the abundance of barcodes linked to each CAR ICD combination. We hypothesized that any remaining CAR-T cells at the tumor site and spleen would represent T cells with long-term persistence elicited by antigen-specific CAR ICD signaling. 103 novel CAR constructs were synthesized based on barcode enrichment from both in vitro and in vivo pooled screens, and we validated their anti-tumor function in a serial tumor rechallenge assay.

Ginkgo CAR library has 10,000 novel ICD combinations

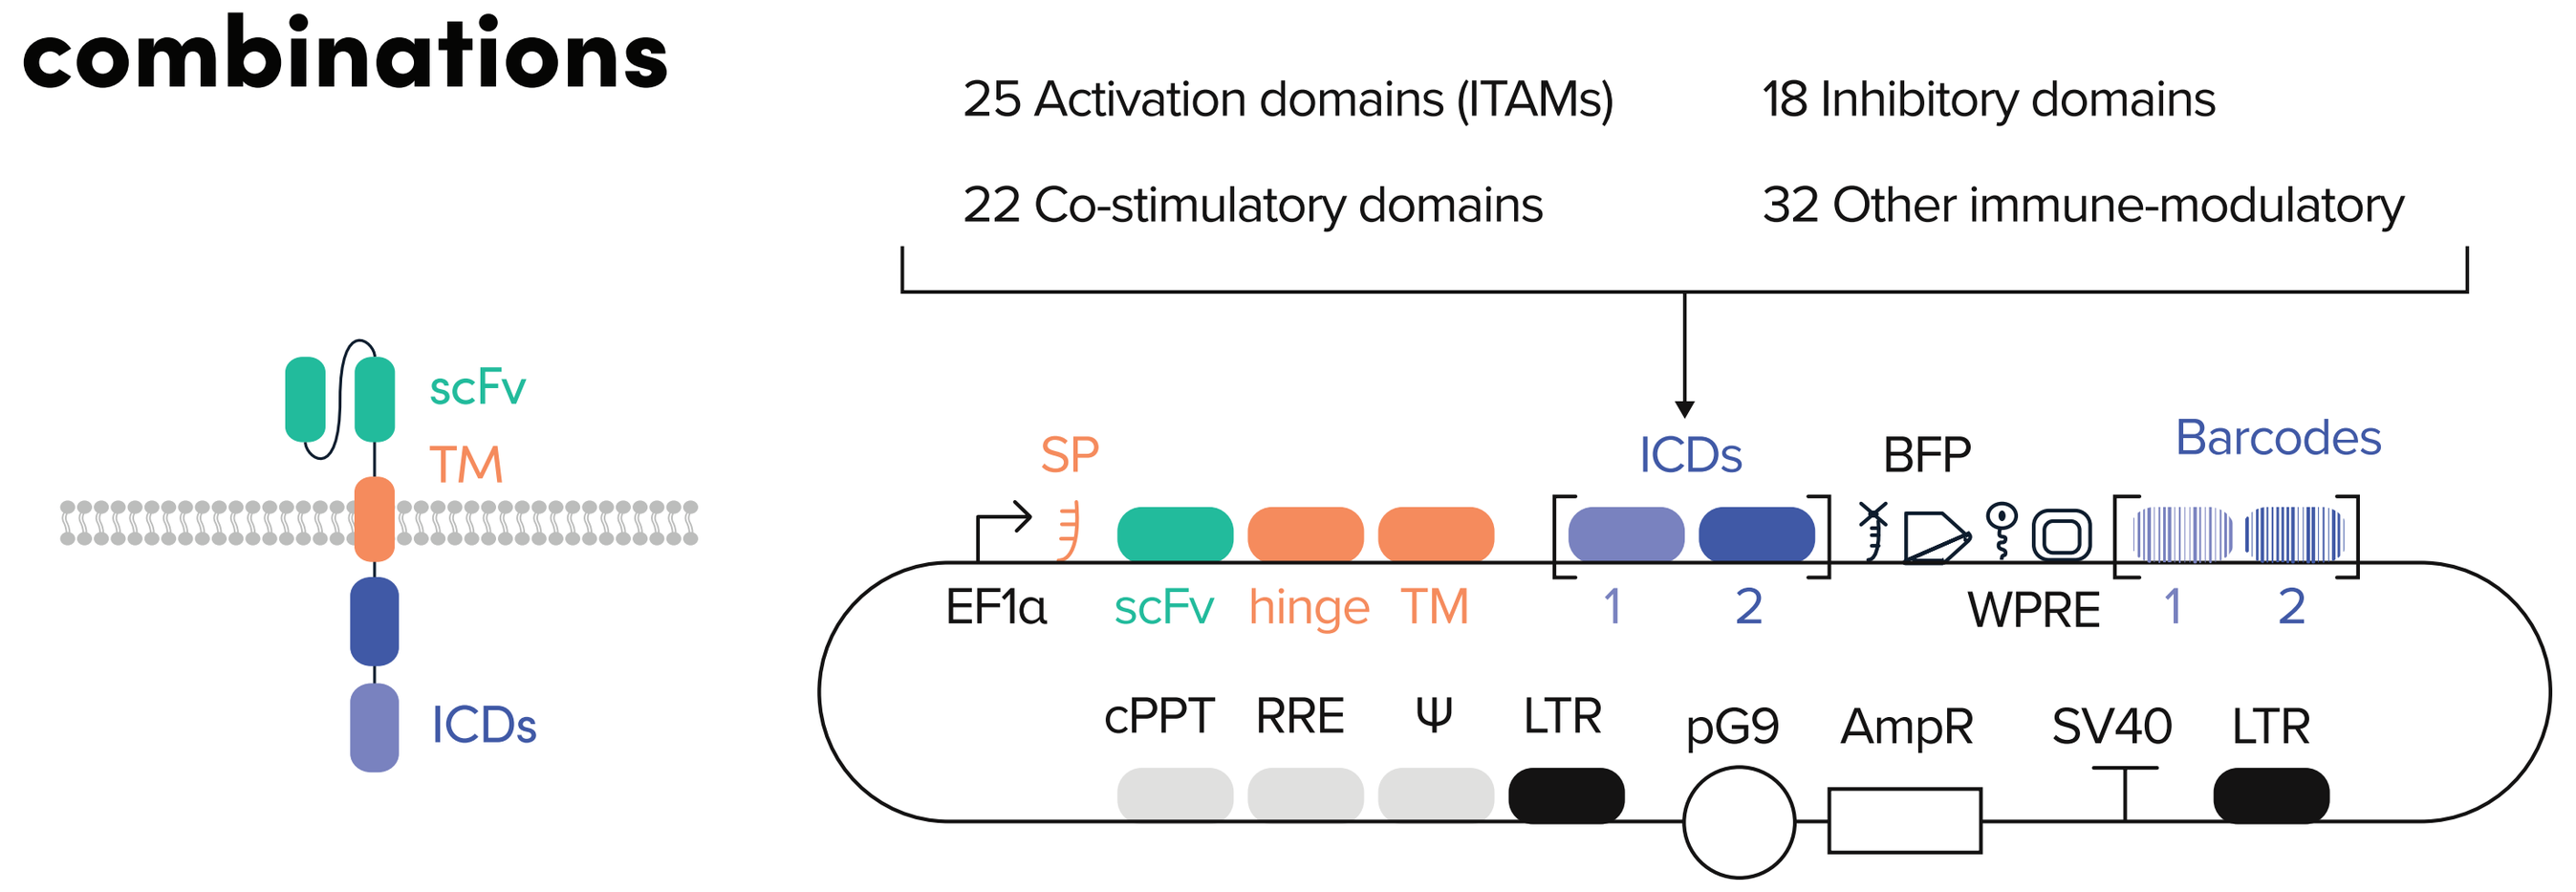
25 Activation domains (ITAMs)
18 Inhibitory domains
22 Co-stimulatory domains
32 Other immune-modulatory
scFv
TM
ICDs
SP
ICDs
BFP
Barcodes
EF1α
scFv
hinge
TM
1
2
WPRE
1
2
cPPT
RRE
Ψ
LTR
pG9
AmpR
SV40
LTR

In vitro/In vivo screen schematics

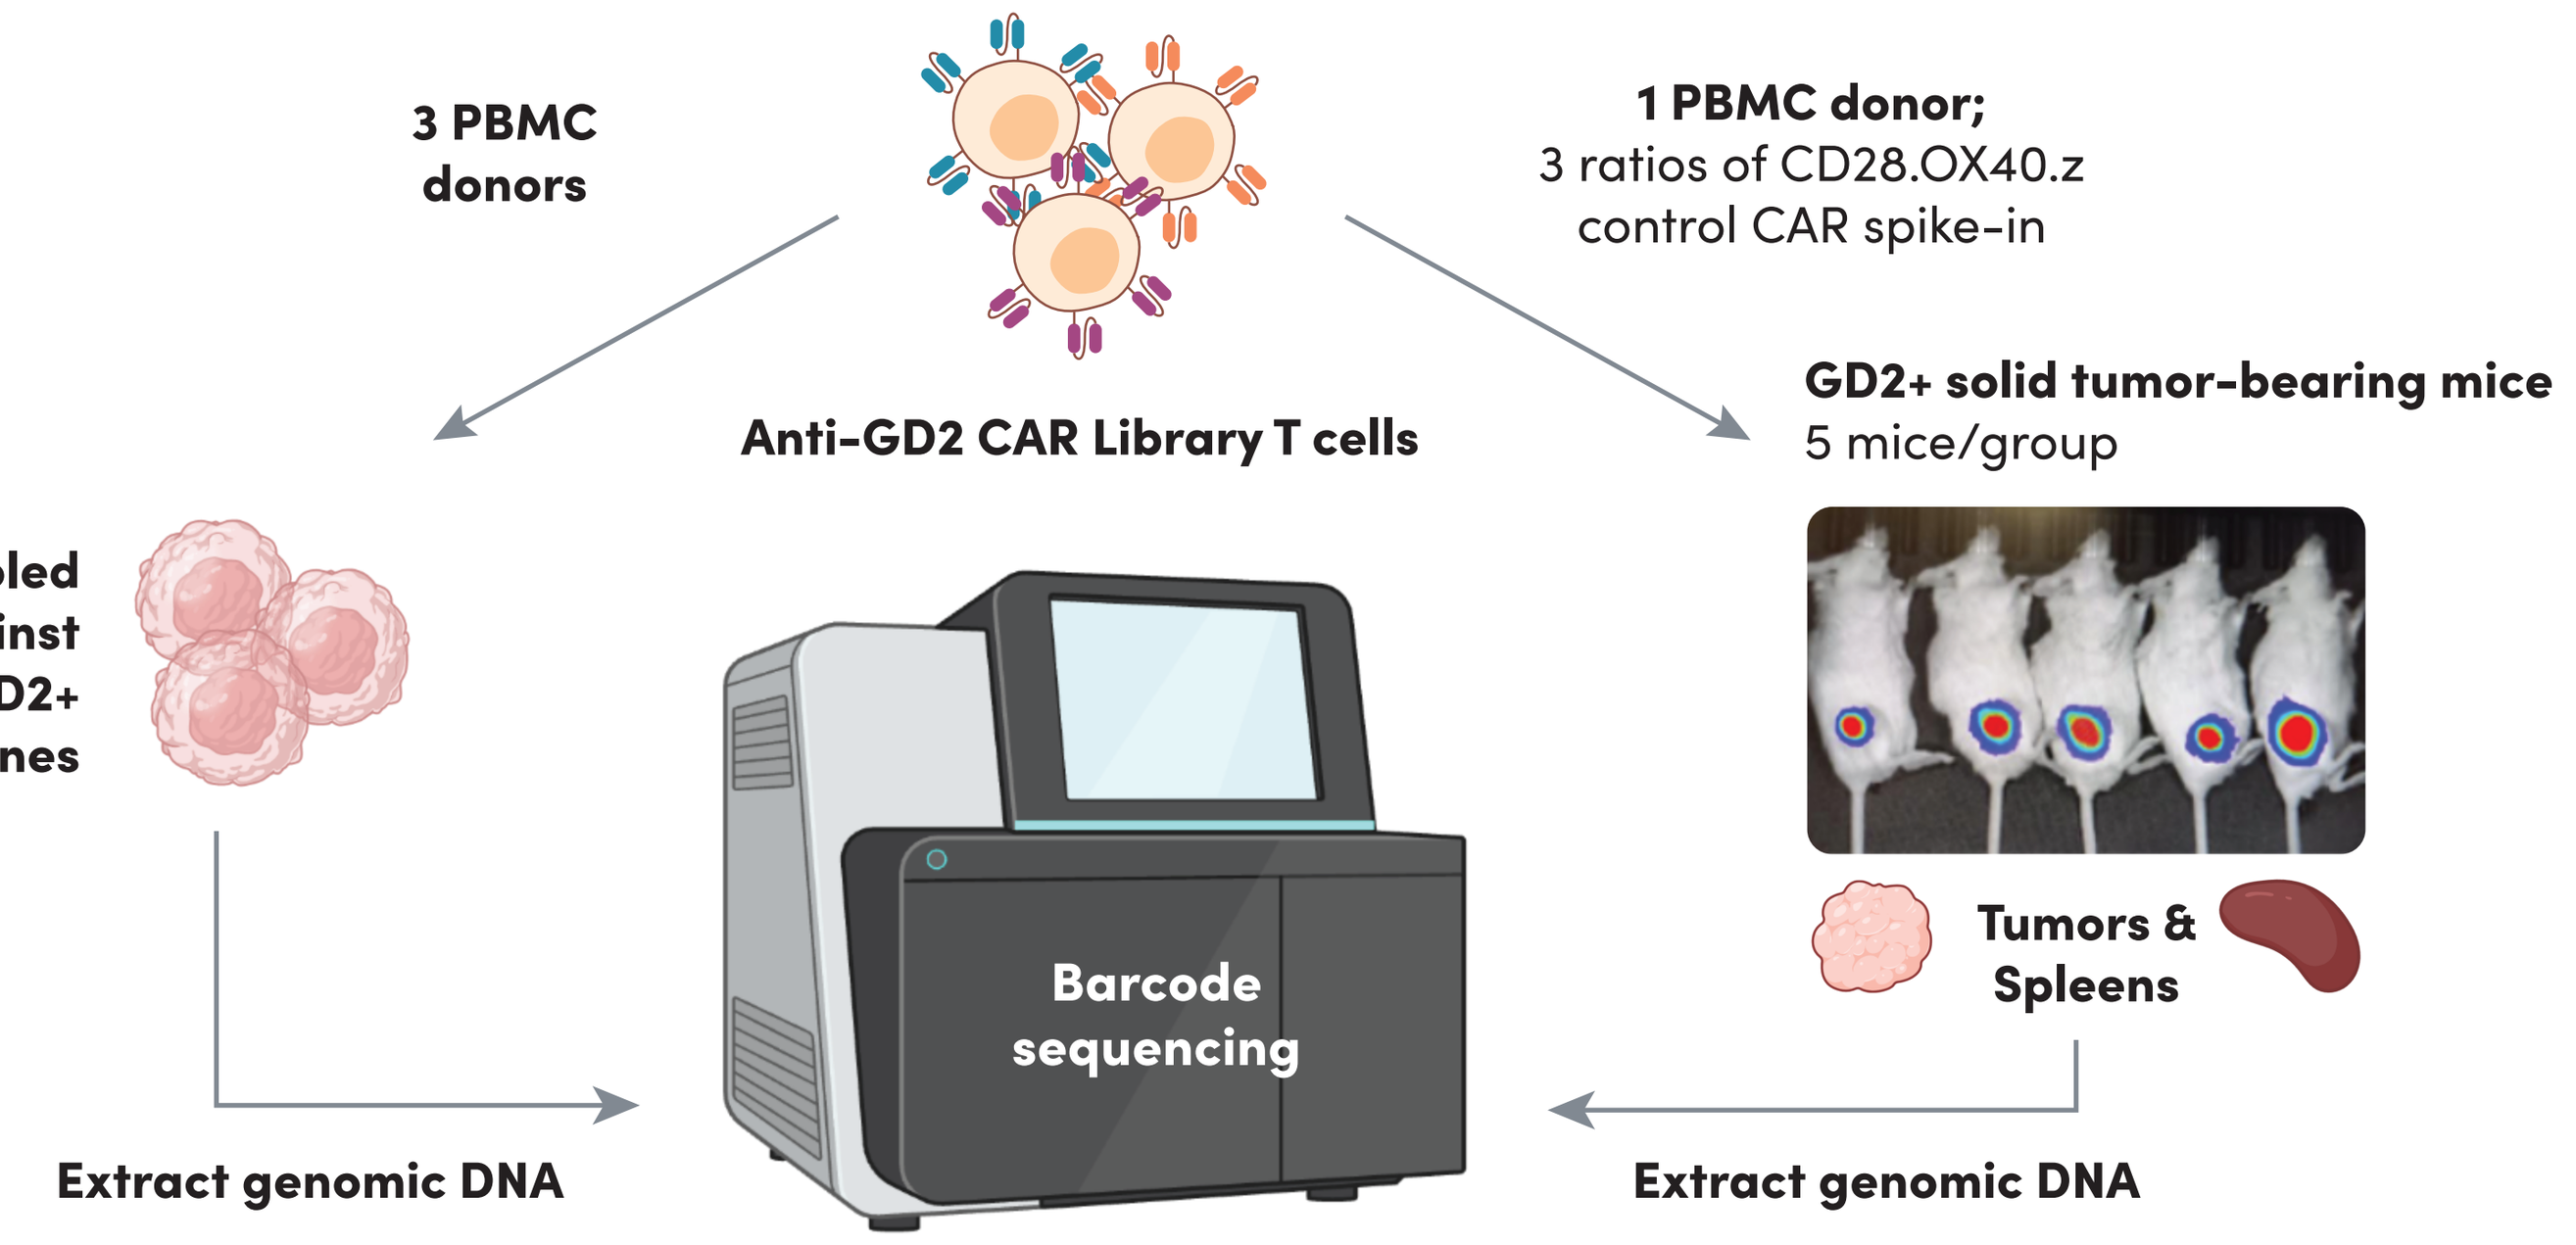
3 PBMC donors
1 PBMC donor;
3 ratios of CD28.OX40.z control CAR spike-in
Anti-GD2 CAR Library T cells
GD2+ solid tumor-bearing mice
5 mice/group
In vitro pooled screen against two GD2+ solid tumor lines
Barcode sequencing
Tumors & Spleens
Extract genomic DNA
Extract genomic DNA

In vitro pooled screen

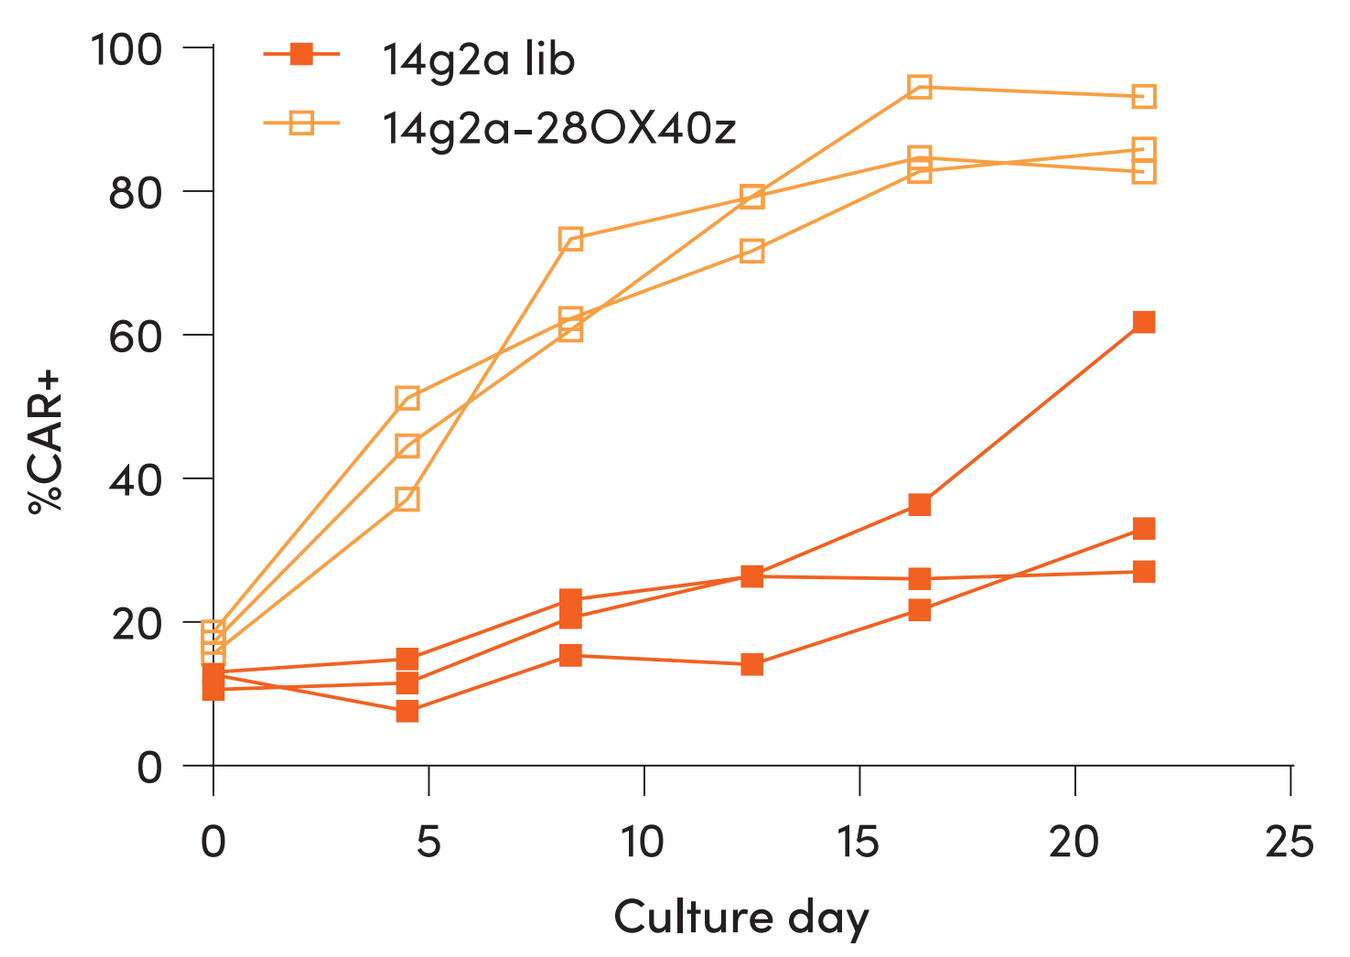
CAR expression, MG-63
14g2a lib
14g2a-28OX40z
%CAR+
Culture day

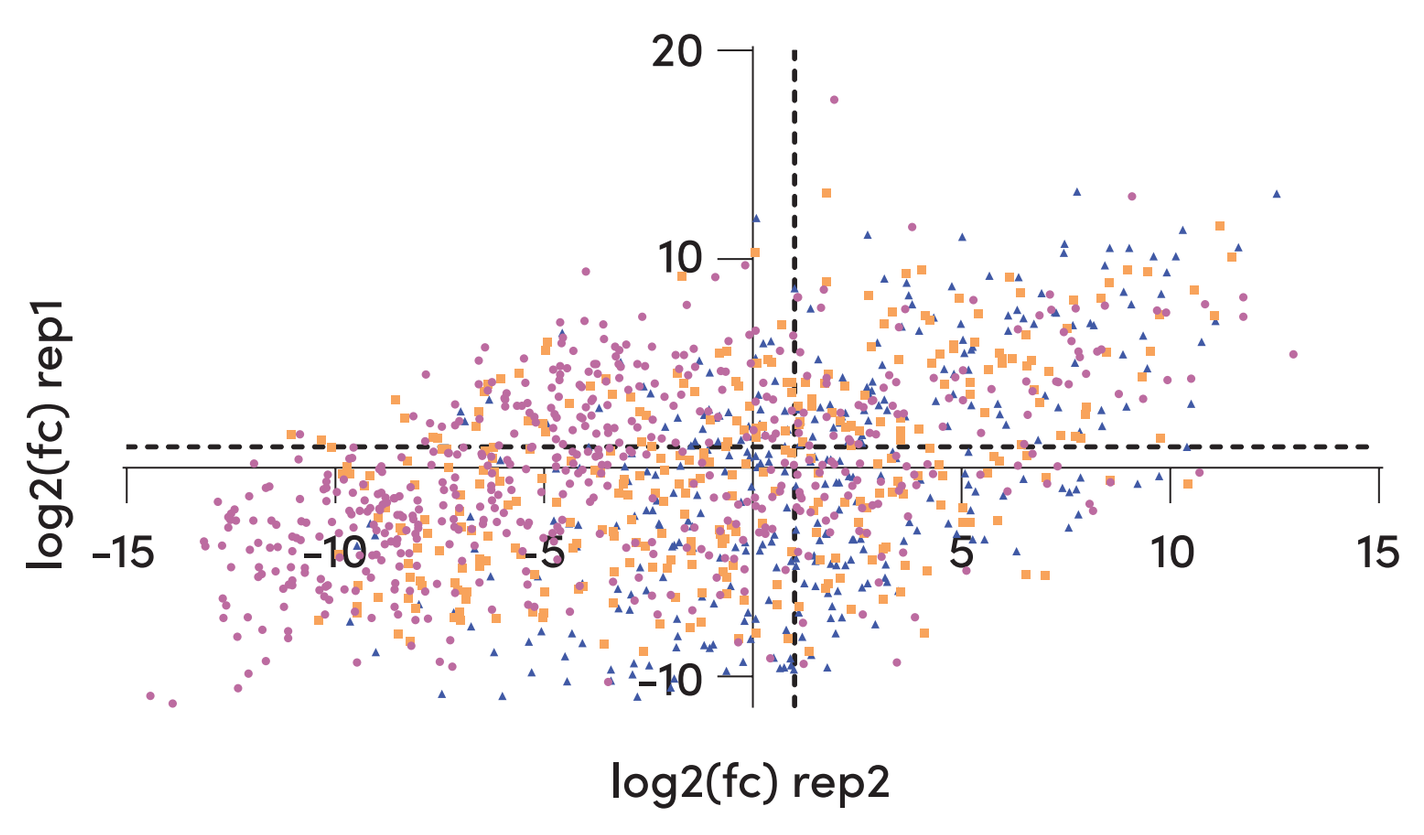
Barcode enrichment (MG-63)
log2(fc) rep1
log2(fc) rep2

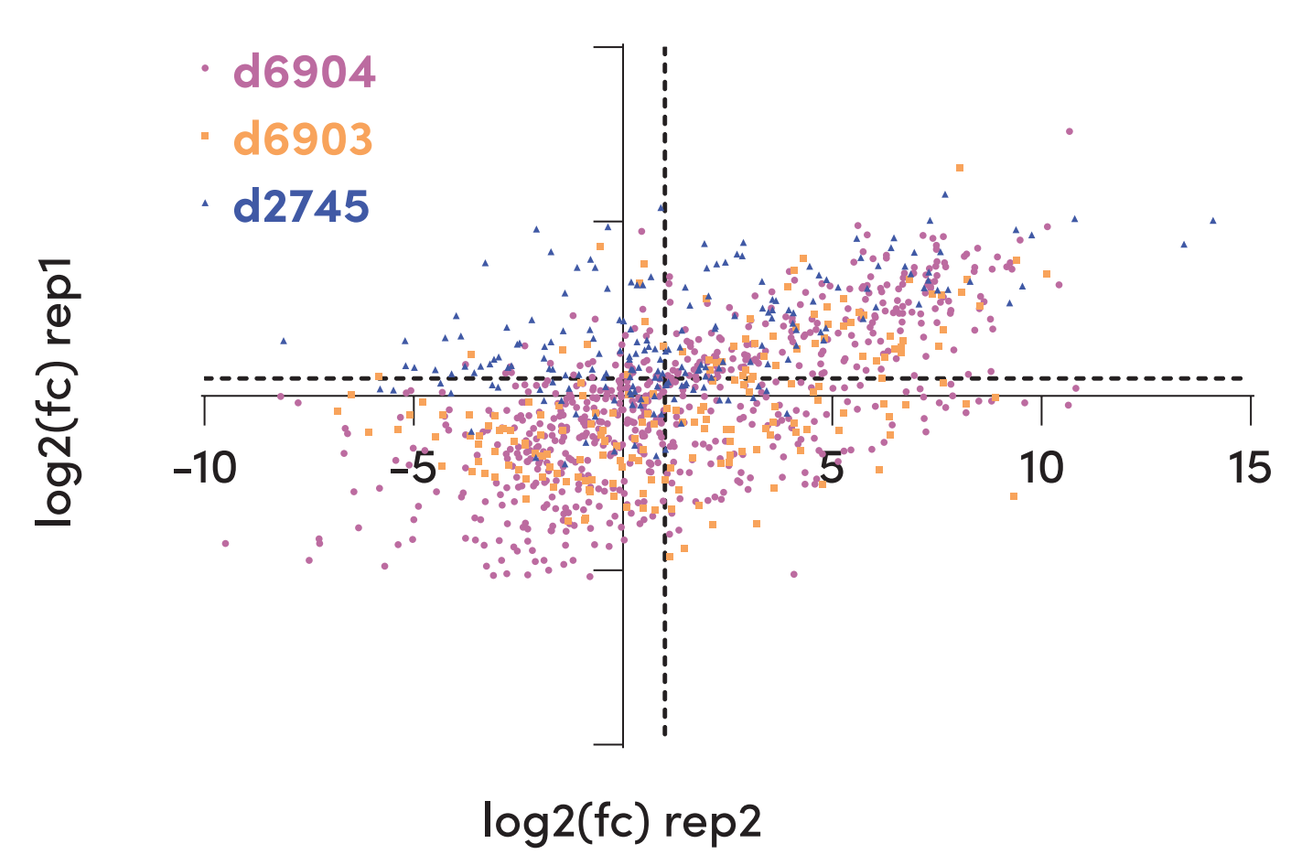
Barcode enrichment (SK-N-AS)
d6904
d6903
d2745
log2(fc) rep1
log2(fc) rep2

In vivo pooled screen

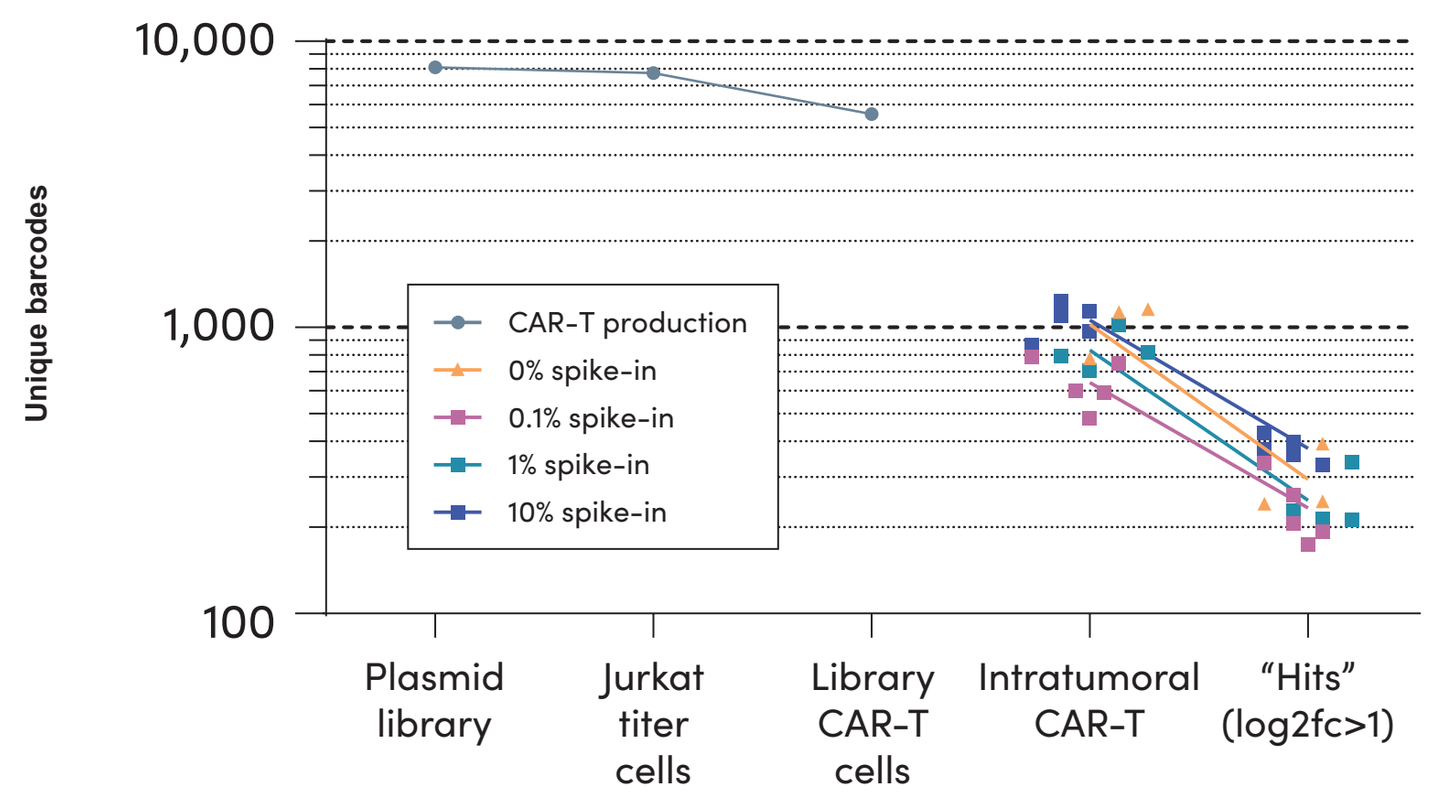
Library diversity throughout in vivo pooled screen
Unique barcodes
CAR-T production
0% spike-in
0.1% spike-in
1% spike-in
10% spike-in
Plasmid library
Jurkat titer cells
Library CAR-T cells
Intratumoral CAR-T
"Hits" (log2fc>1)

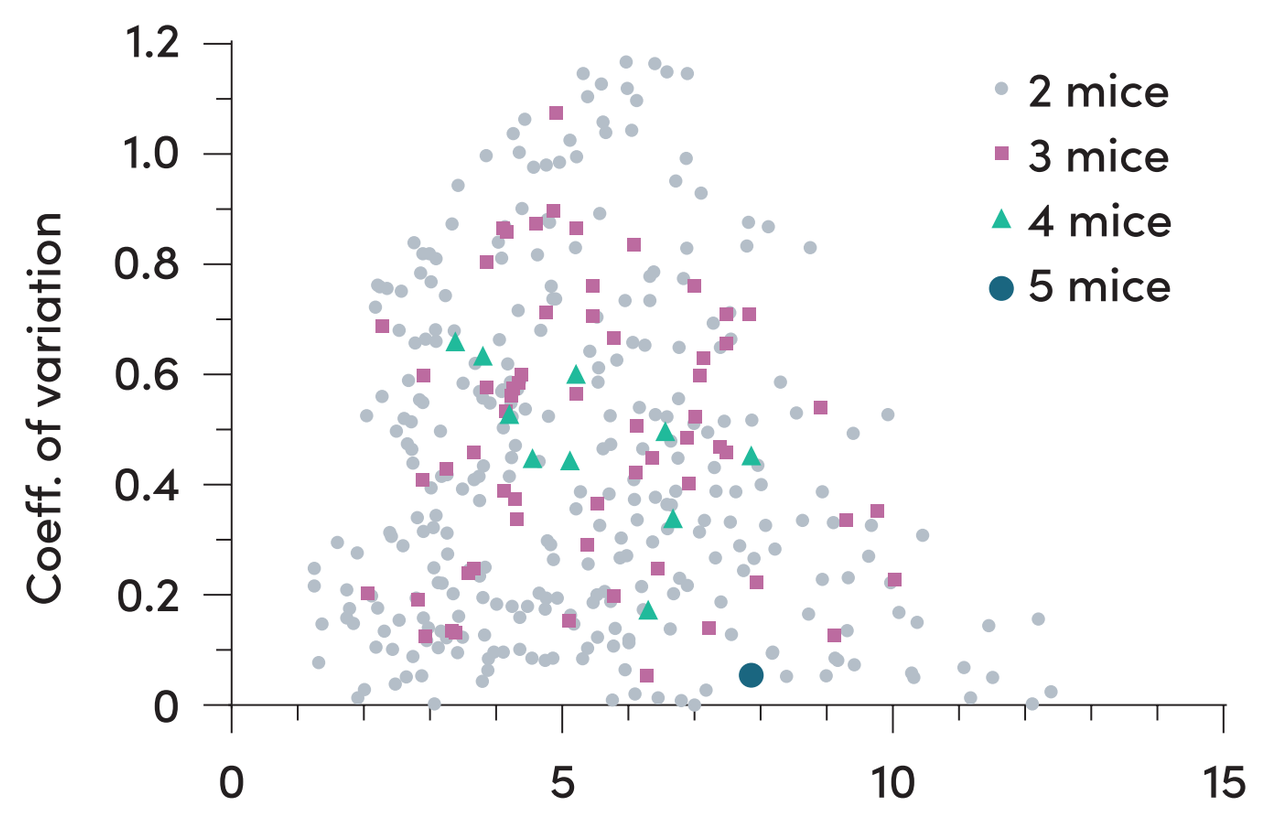
Enriched barcodes found in ≥2 mouse tumors
Coeff. of variation
2 mice
3 mice
4 mice
5 mice

Relative frequency of ICD appearance among enriched barcodes from in vitro and in vivo pooled screens

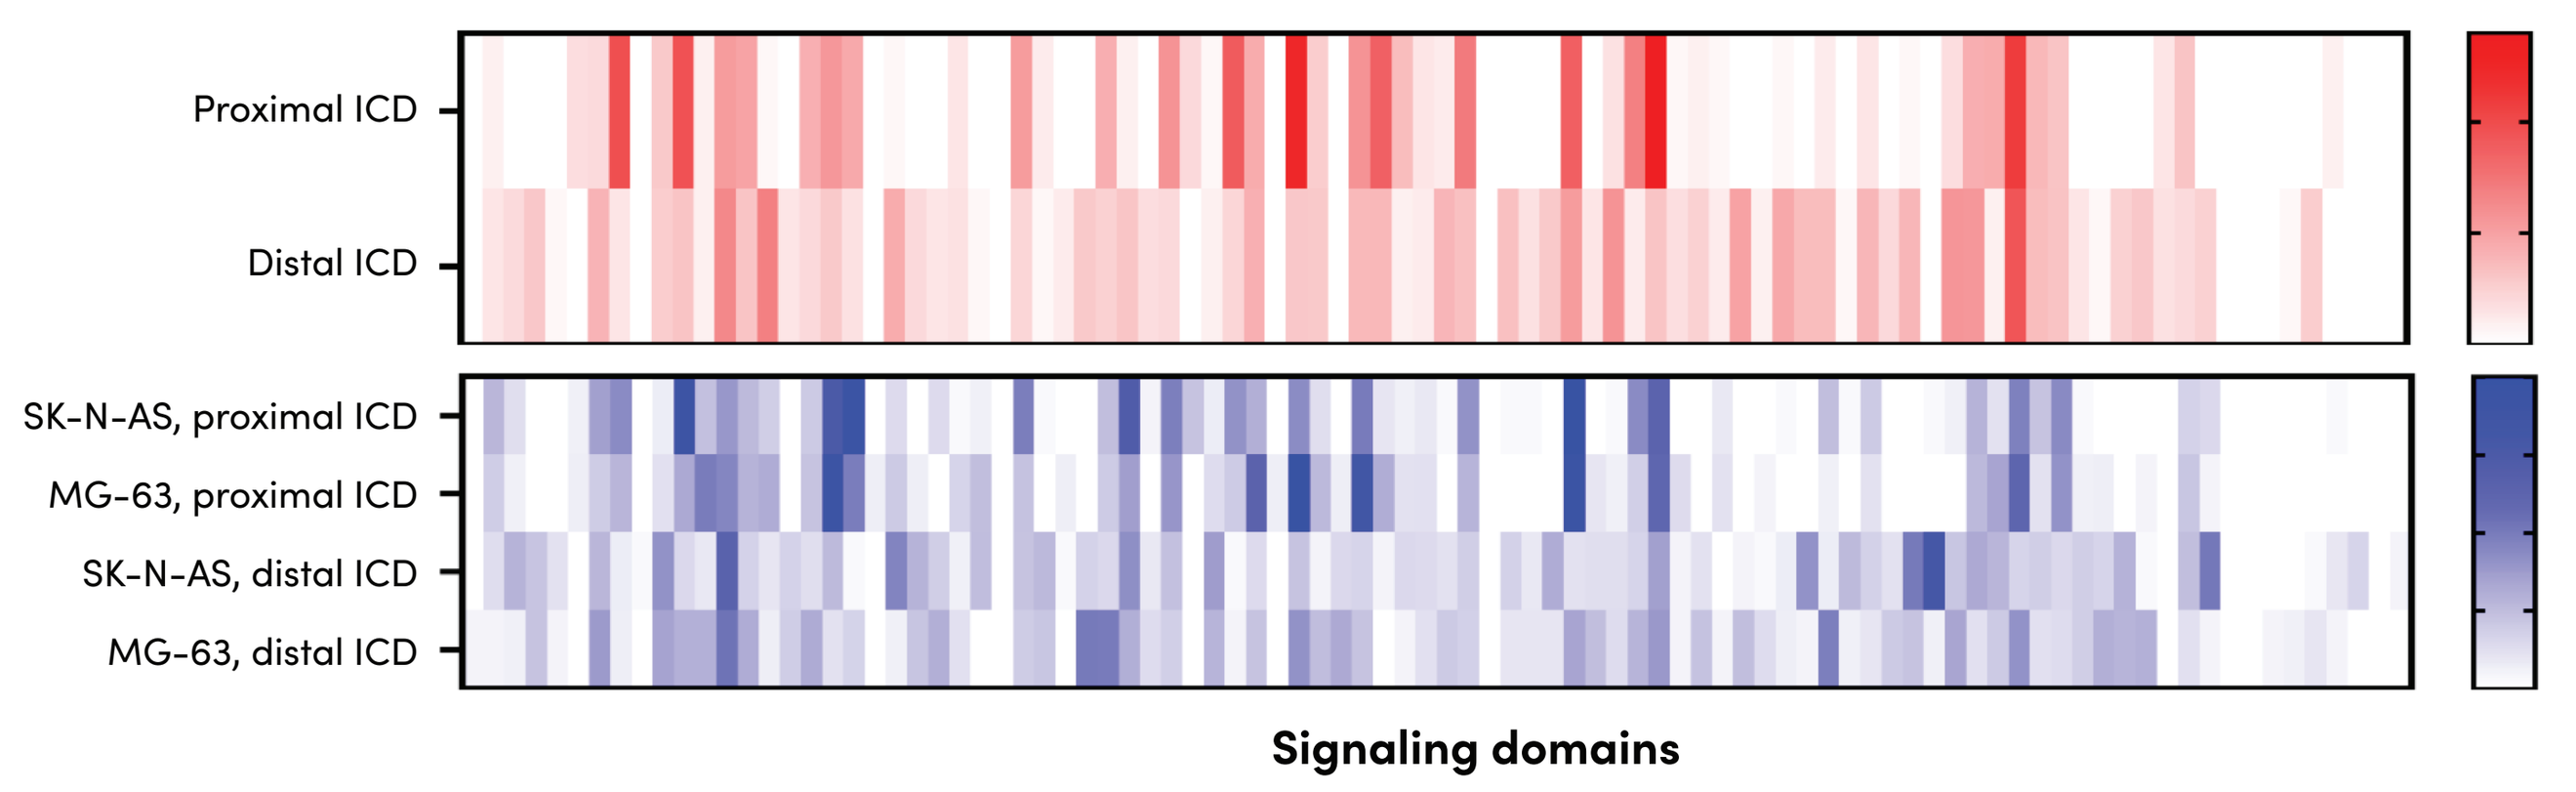
Proximal ICD
Distal ICD
SK-N-AS, proximal ICD
MG-63, proximal ICD
SK-N-AS, distal ICD
MG-63, distal ICD
Signaling domains

Methodology for selecting in vitro pooled screening hits

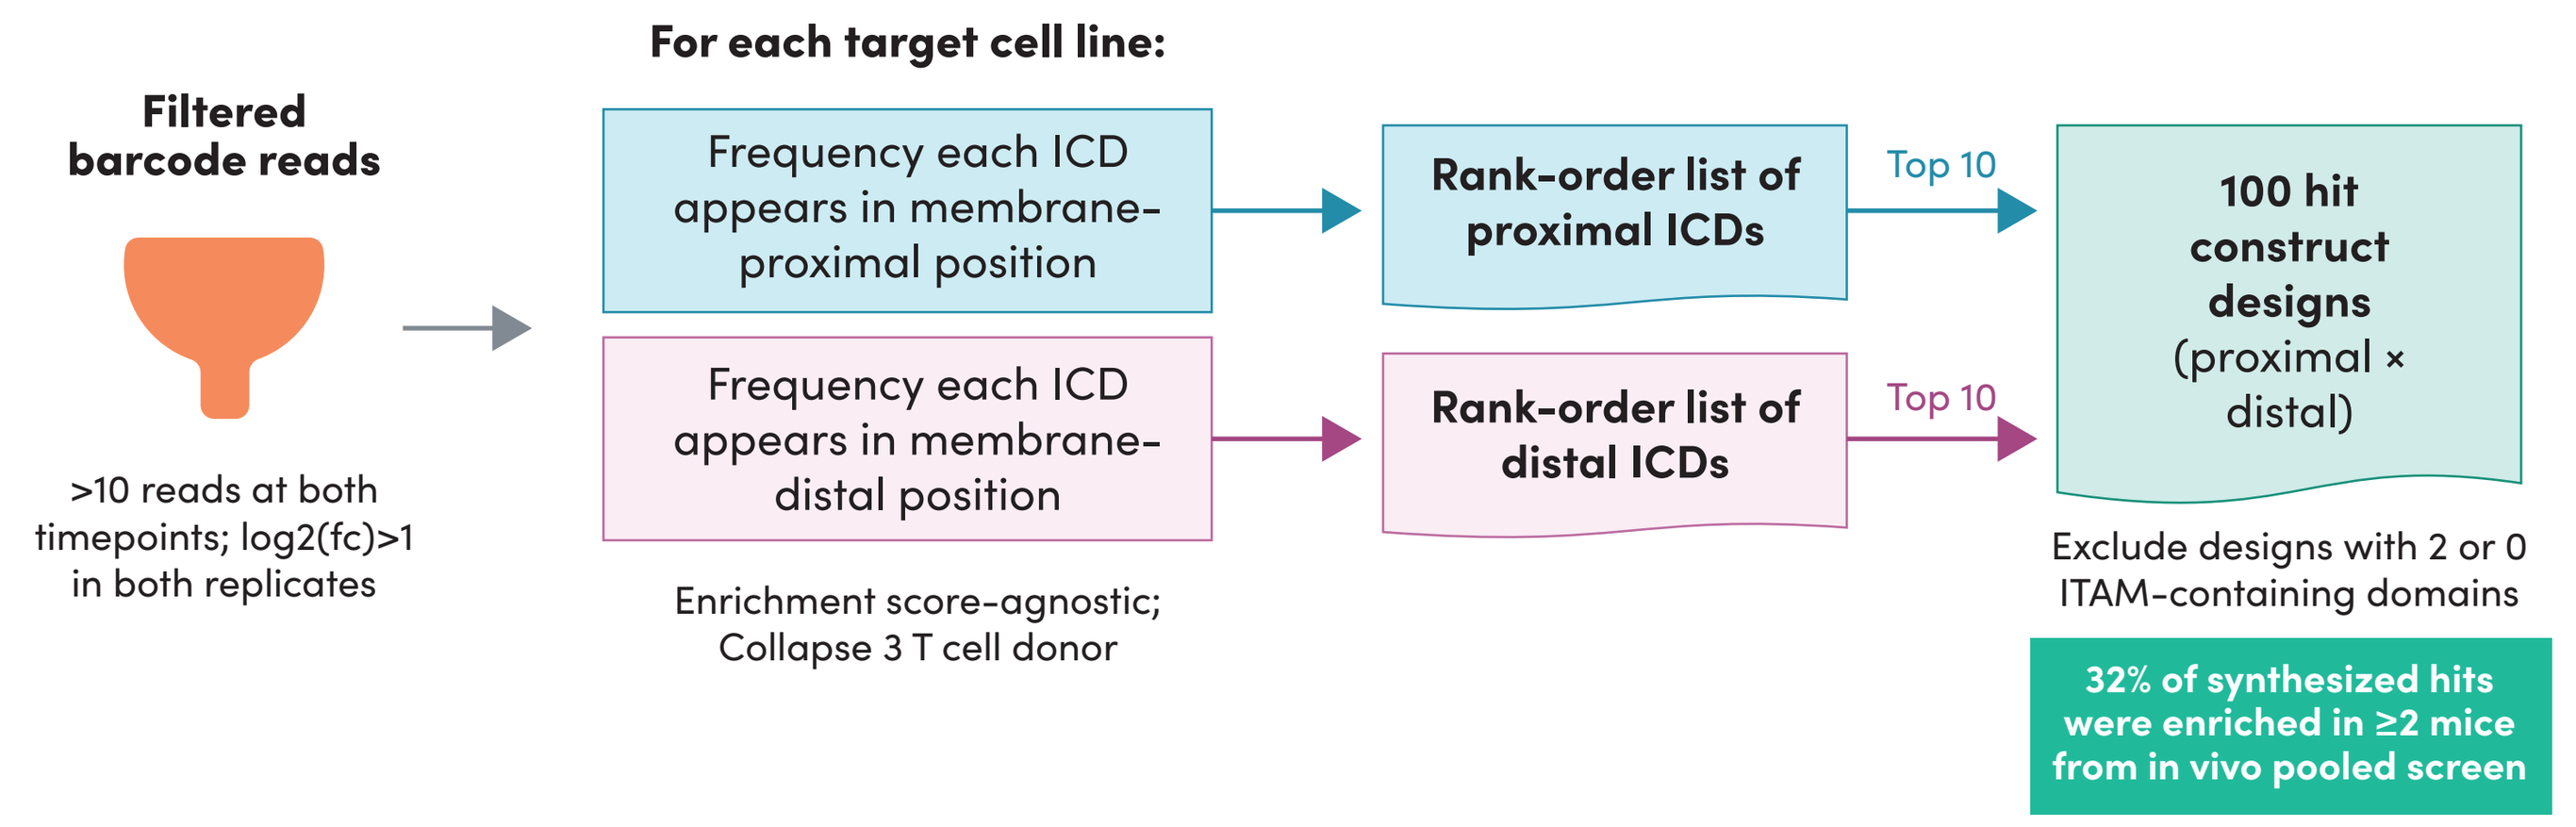
For each target cell line:
Filtered barcode reads
>10 reads at both timepoints; log2(fc)>1 in both replicates
Frequency each ICD appears in membrane-proximal position
Frequency each ICD appears in membrane-distal position
Enrichment score-agnostic; Collapse 3 T cell donor
Rank-order list of proximal ICDs
Rank-order list of distal ICDs
Top 10
Top 10
100 hit construct designs (proximal × distal)
Exclude designs with 2 or 0 ITAM-containing domains
32% of synthesized hits were enriched in ≥2 mice from in vivo pooled screen

Many novel CARs perform better than clinical anti-GD2 CAR design against two GD2+ tumor cell lines with differing antigen densities

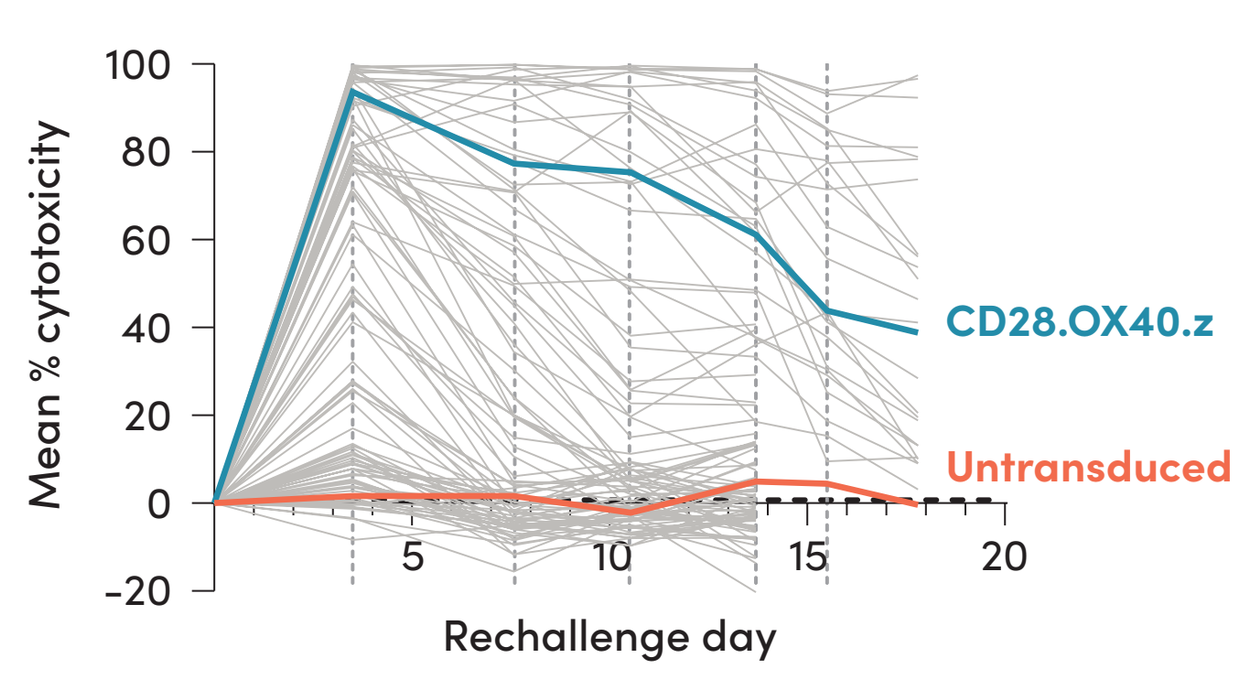
Serial killing against MG-63
Mean % cytotoxicity
CD28.OX40.z
Untransduced
Rechallenge day

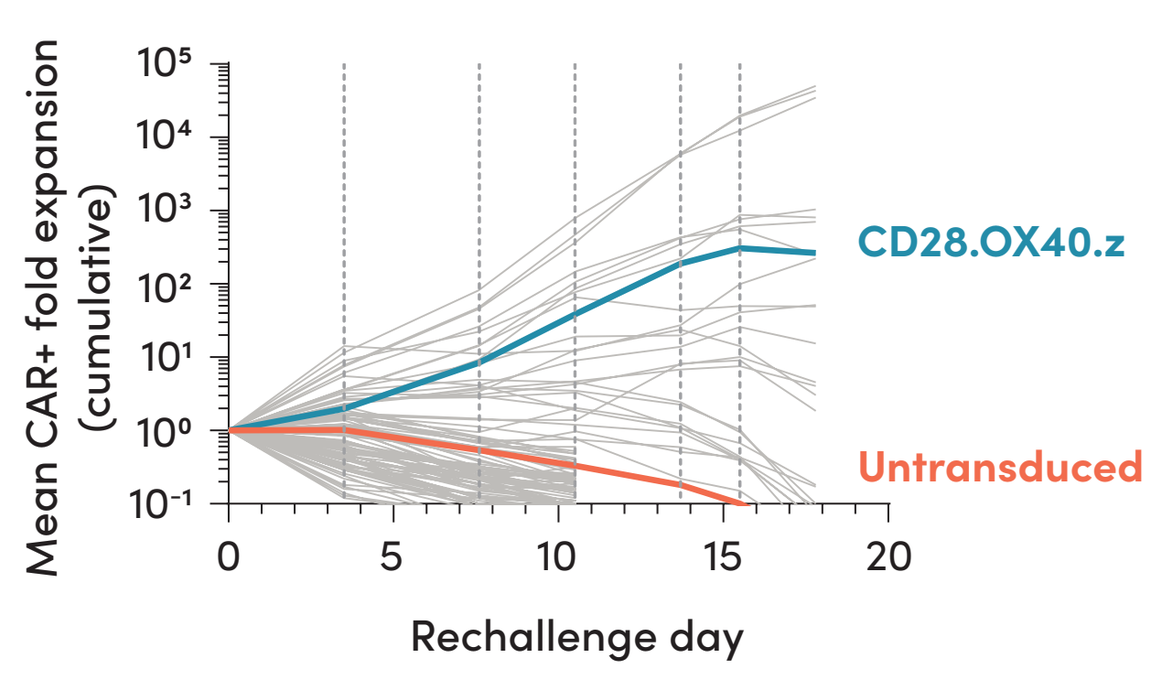
CAR+ expansion against MG-63
Mean CAR+ fold expansion (cumulative)
CD28.OX40.z
Untransduced
Rechallenge day

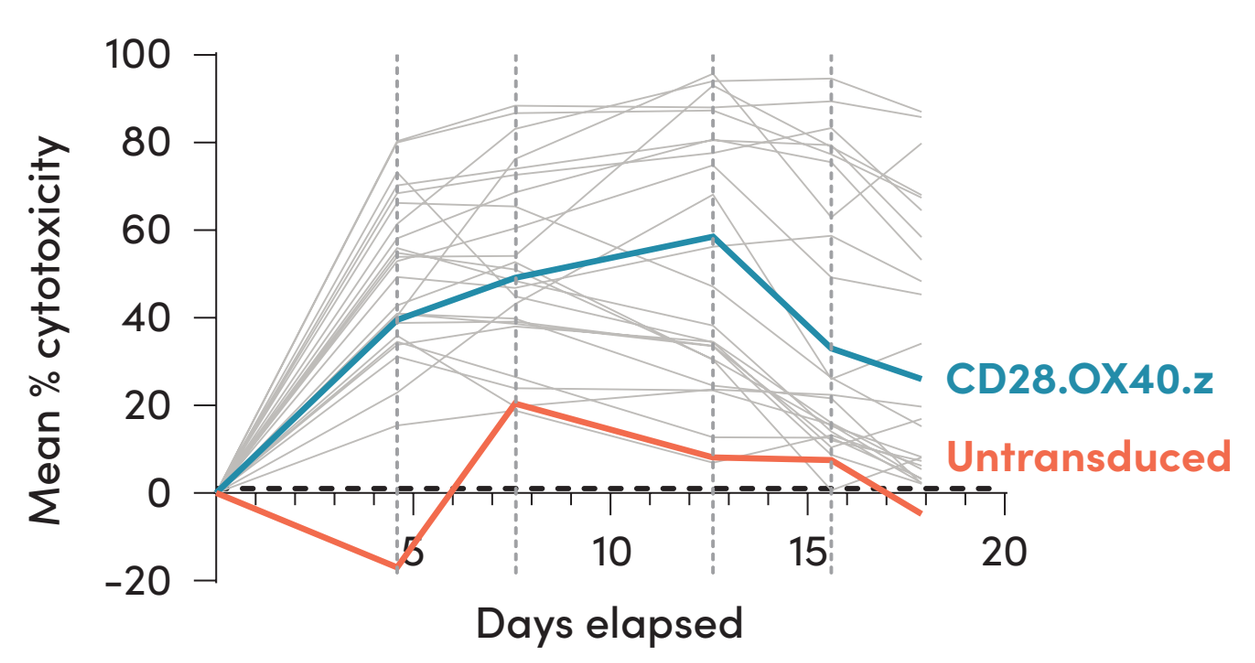
Serial killing against SK-N-AS
Mean % cytotoxicity
CD28.OX40.z
Untransduced
Days elapsed

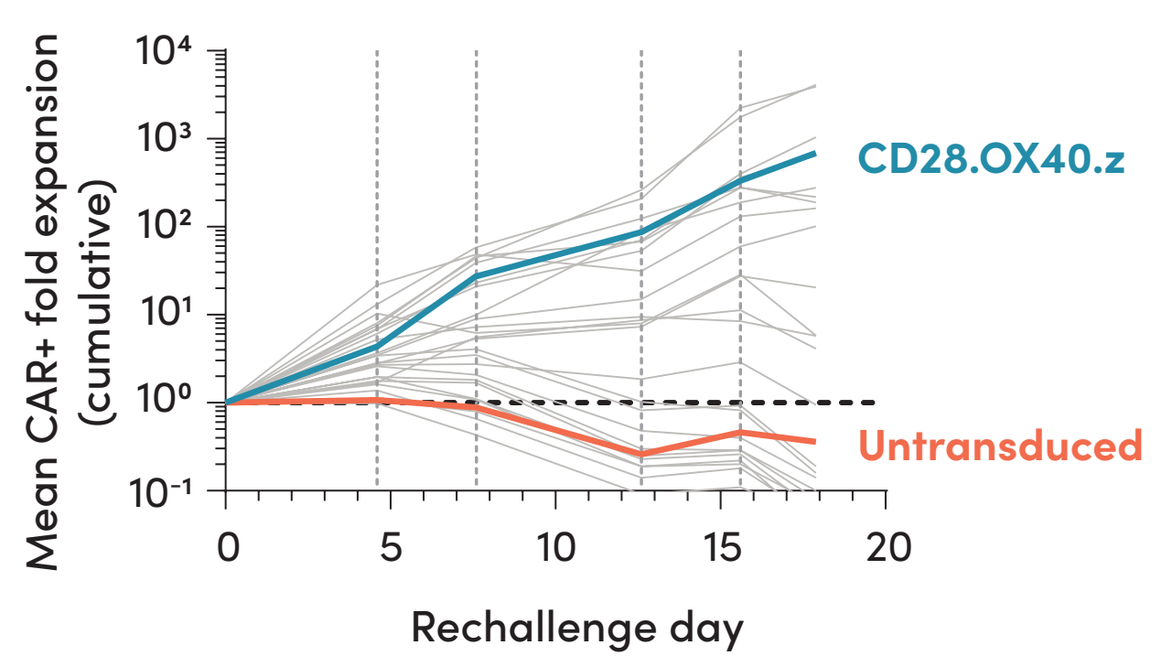
CAR+ expansion against SK-N-AS
Mean CAR+ fold expansion (cumulative)
CD28.OX40.z
Untransduced
Rechallenge day

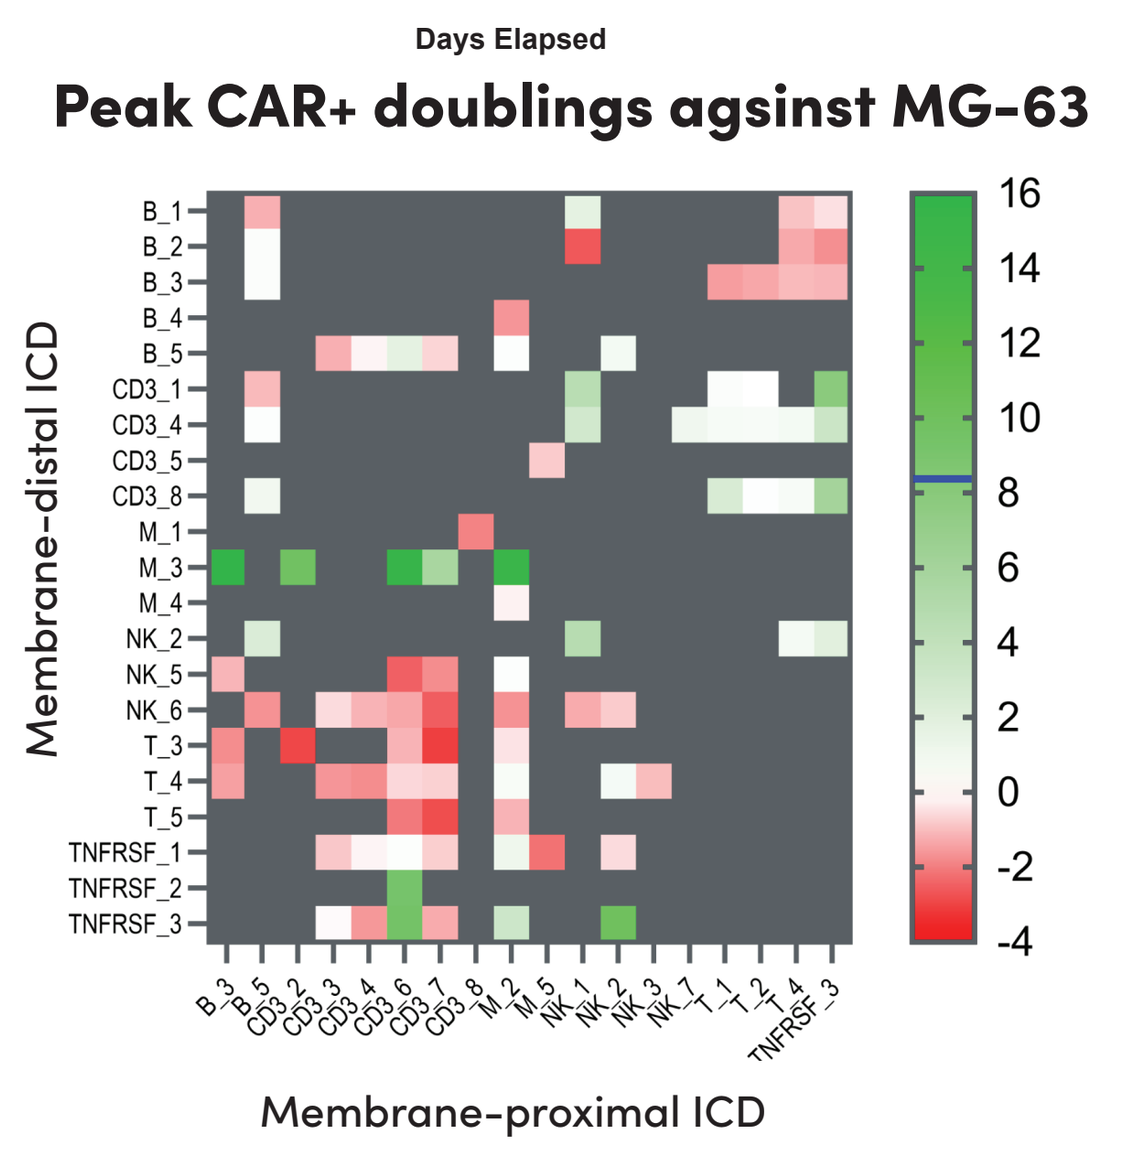
Days Elapsed
Peak CAR+ doublings agsinst MG-63
Membrane-distal ICD
Membrane-proximal ICD

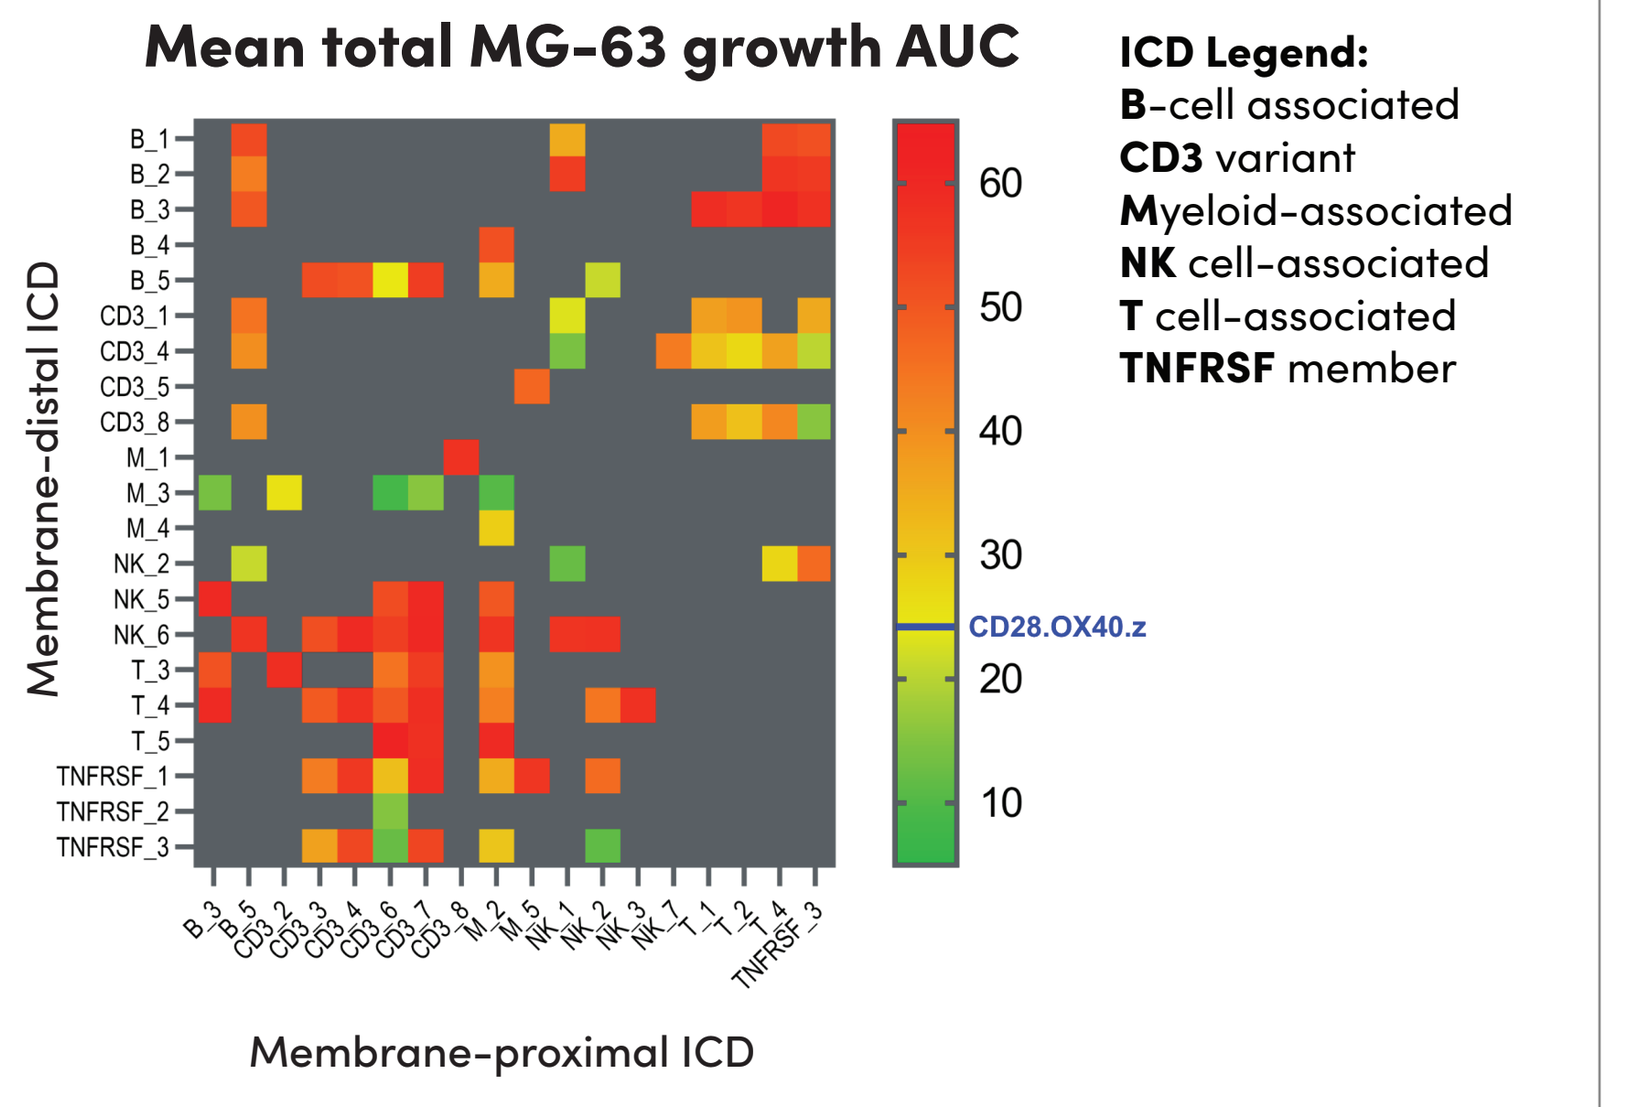
Mean total MG-63 growth AUC
Membrane-distal ICD
Membrane-proximal ICD
CD28.OX40.z
ICD Legend:
B-cell associated
CD3 variant
Myeloid-associated
NK cell-associated
T cell-associated
TNFRSF member

Many novel CARs exhibit better long-term tumor control than clinical CAR, despite less robust proliferation

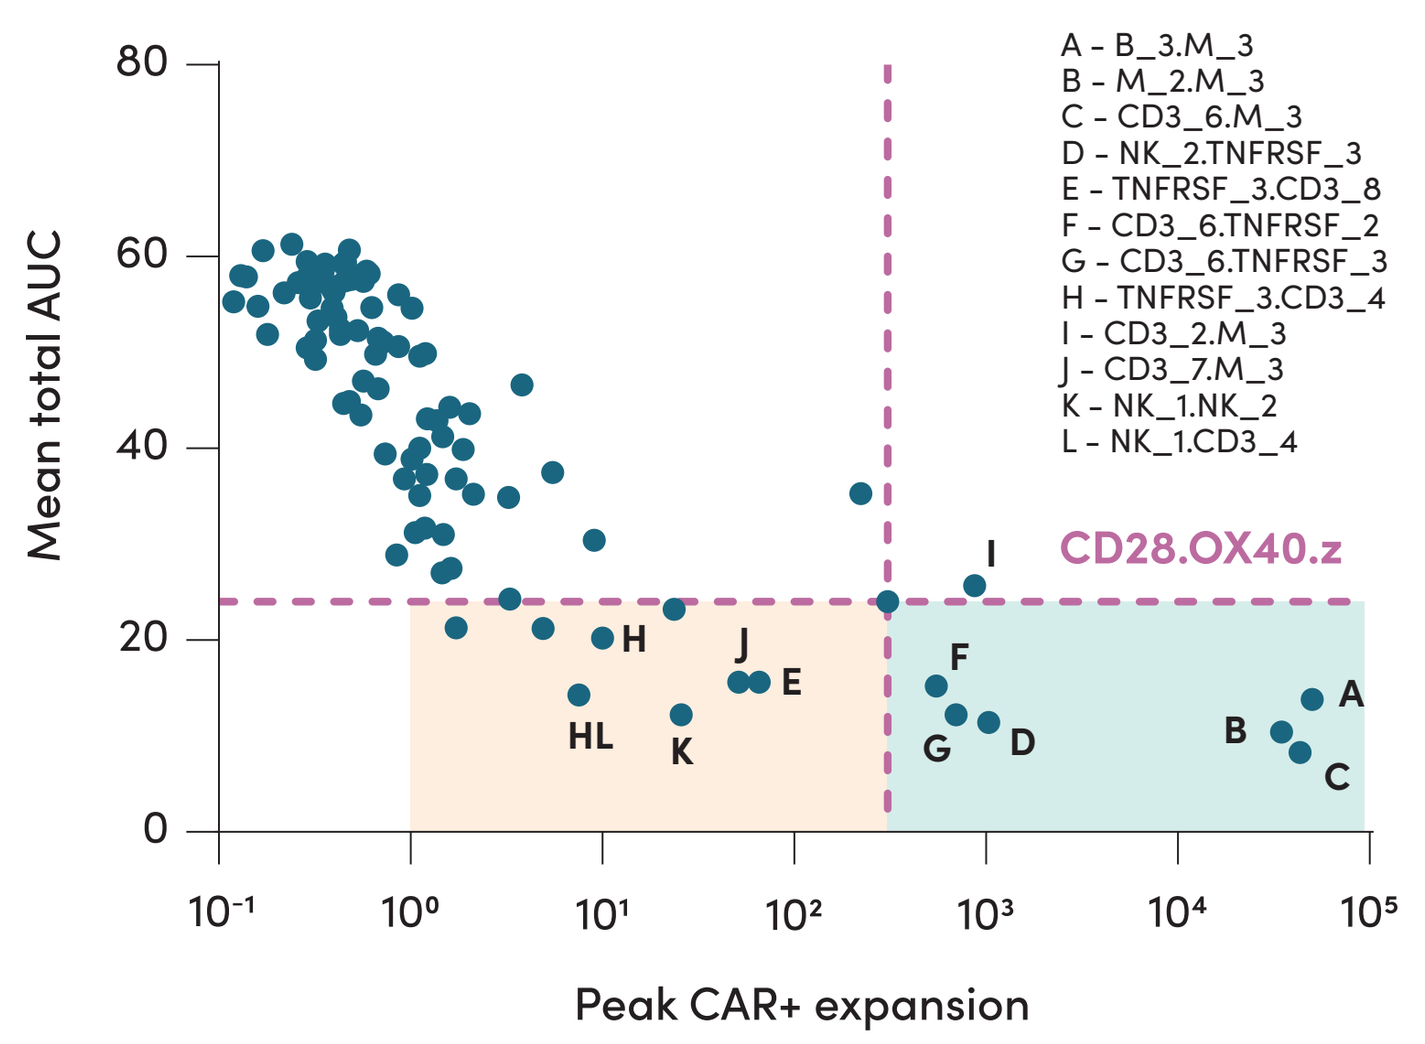
Proliferation vs. killing against MG-63
Mean total AUC
Peak CAR+ expansion
CD28.OX40.z
A - B_3.M_3
B - M_2.M_3
C - CD3_6.M_3
D - NK_2.TNFRSF_3
E - TNFRSF_3.CD3_8
F - CD3_6.TNFRSF_2
G - CD3_6.TNFRSF_3
H - TNFRSF_3.CD3_4
I - CD3_2.M_3
J - CD3_7.M_3
K - NK_1.NK_2
L - NK_1.CD3_4

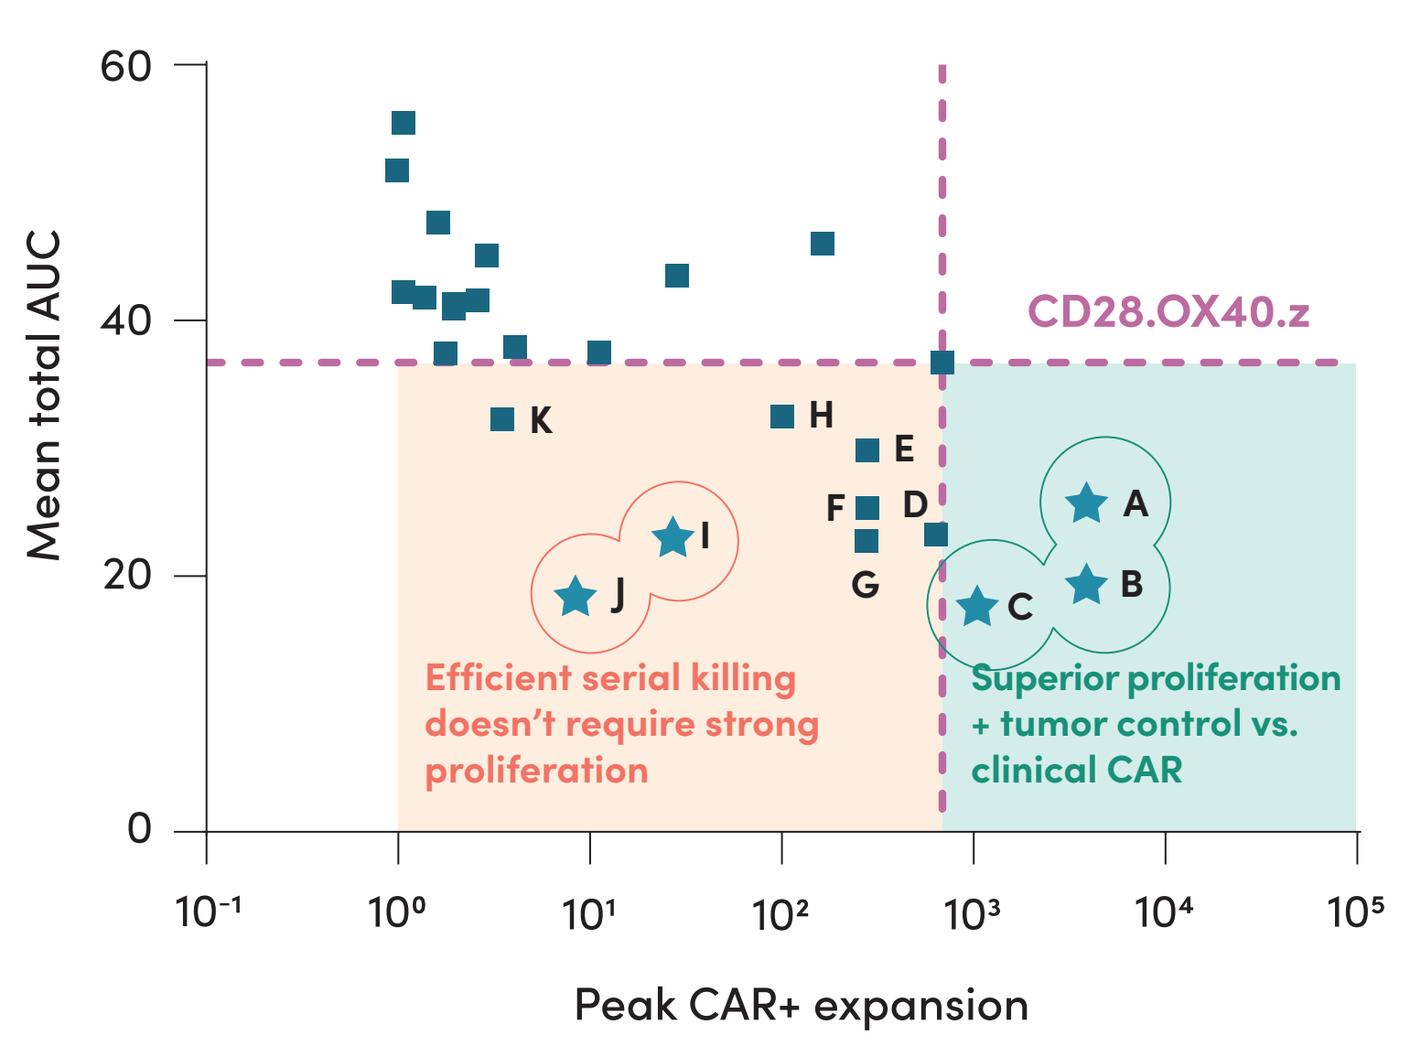
Proliferation vs. killing against SK-N-AS
Mean total AUC
Peak CAR+ expansion
CD28.OX40.z
Efficient serial killing doesn't require strong proliferation
Superior proliferation + tumor control vs. clinical CAR

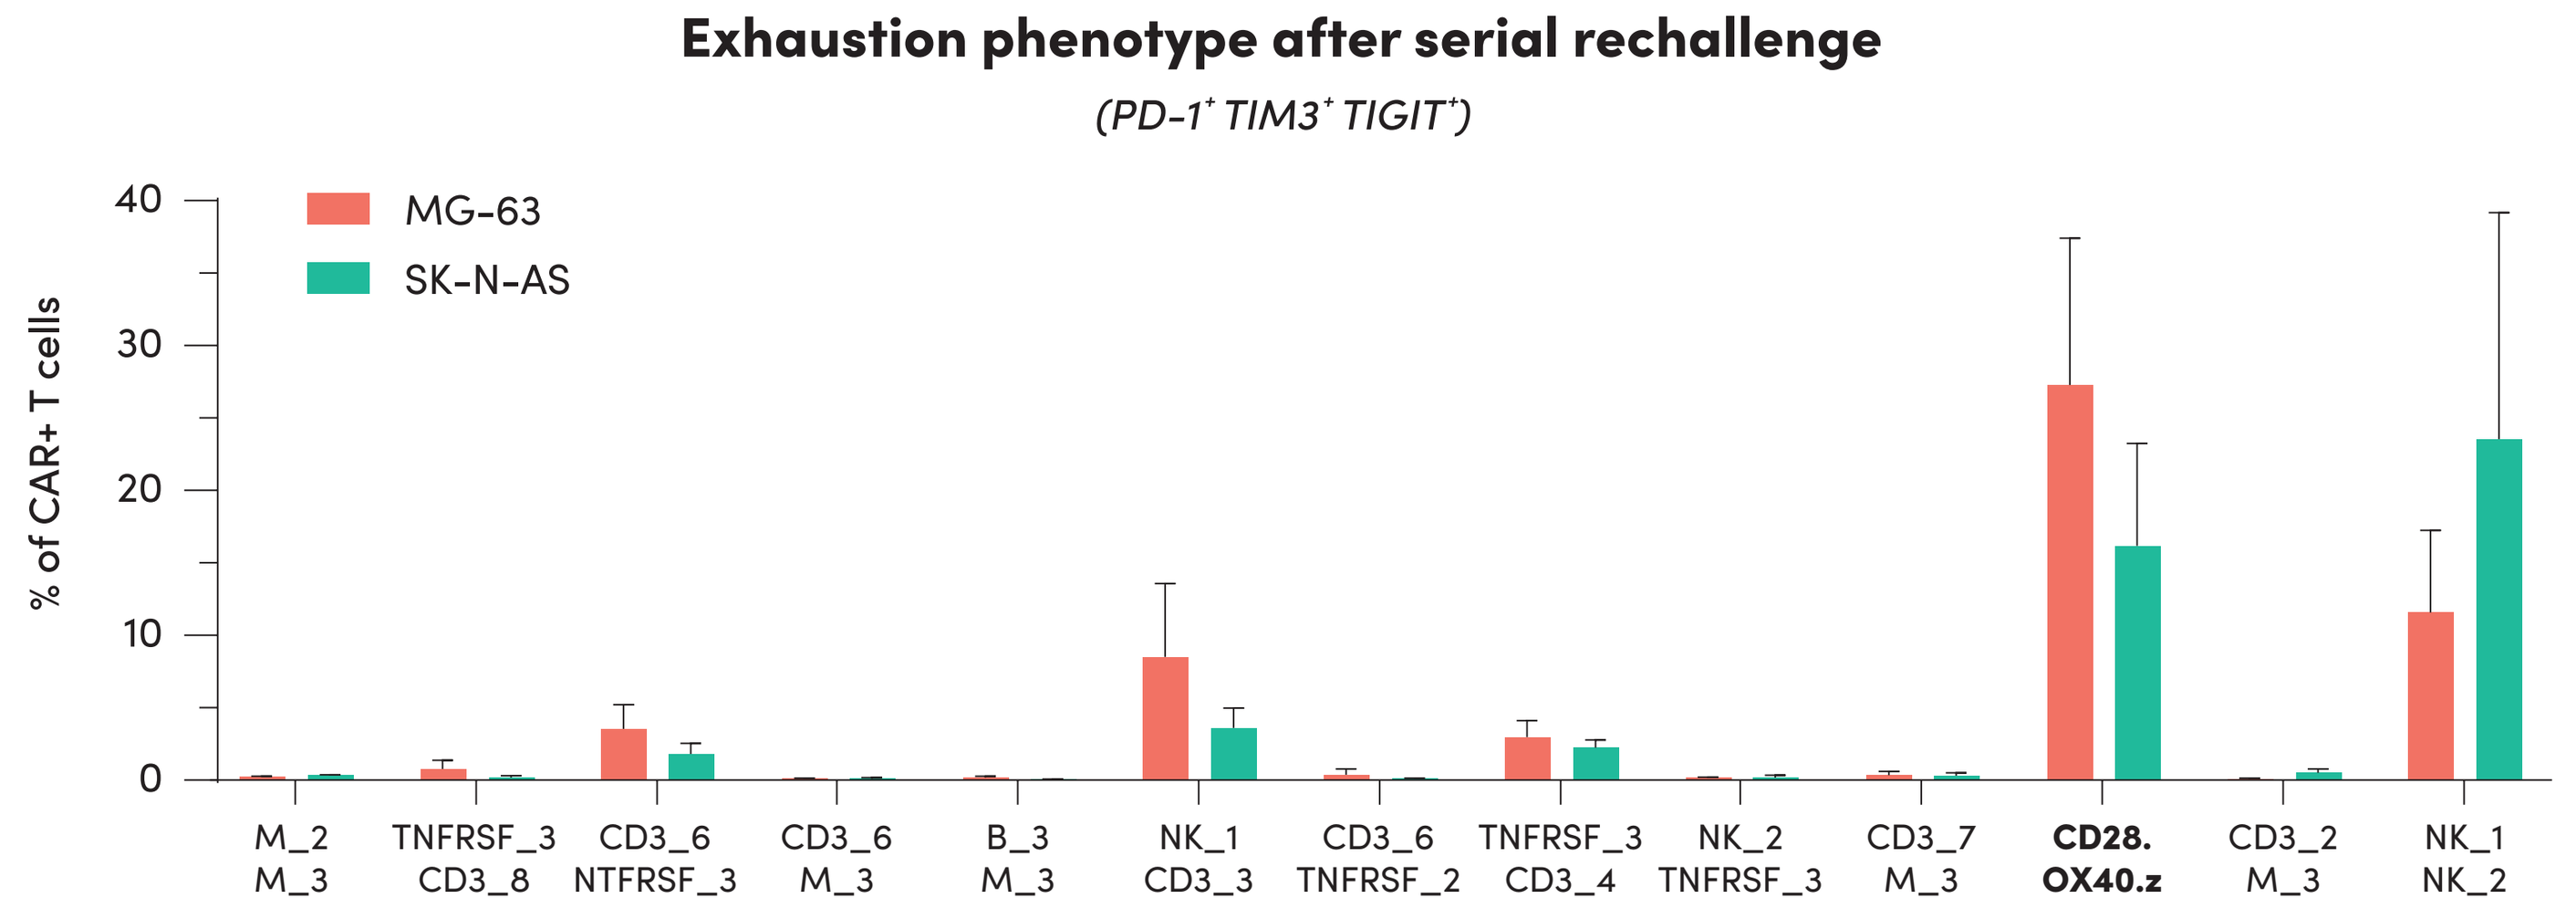
Exhaustion phenotype after serial rechallenge
(PD-1+ TIM3+ TIGIT+)
% of CAR+ T cells
MG-63
SK-N-AS
M_2 M_3
TNFRSF_3 CD3_8
CD3_6 NTFRSF_3
CD3_6 M_3
B_3 M_3
NK_1 CD3_3
CD3_6 TNFRSF_2
TNFRSF_3 CD3_4
NK_2 TNFRSF_3
CD3_7 M_3
CD28. OX40.z
CD3_2 M_3
NK_1 NK_2

Conclusions

Here we have demonstrated that both in vitro and in vivo pooled screening can discover novel ICD combinations with superior function against solid tumors compared to current clinical CAR designs. In particular, in vivo pooled screening allowed for high coverage of library diversity, and hundreds of unique enriched barcodes were recovered from digested mouse tumors. Many barcodes were consistently and highly enriched across multiple replicate mice, increasing confidence in the veracity of these hits. To overcome high donor-to-donor variability and mitigate false positives in our in vitro pooled screen, we employed a novel approach wherein hit designs were generated based on frequency of ICD appearance among enriched barcodes.

Small-scale arrayed screening identified numerous novel CAR designs with superior proliferation, cytotoxicity, and phenotype after nearly three weeks of chronic antigen stimulation. ICD identities among the best-performing designs indicate rules for superior CAR architecture; namely, membrane-proximal placement of ITAM-containing domains and inclusion of non-T cell-derived immune signaling motifs. Further development of an in vivo screening platform in humanized mice utilizing multiple PBMC donors could be a promising next step to discovering cell therapy products with strong translational potential.